Laboratory Performance of the DopaFuse® Delivery System

R. Draper[1], J. Harmon[1], A. Heller[1,2], B. Heller[1], E. Heller[1], C. King[1], T. Lau[1], P. Plante[1], J. Spiridigliozzi[1]
1. SynAgile Corporation, Wilson, WY, USA 2. University of Texas at Austin, USA

Objective

SynAgile has developed the DopaFuse Delivery System to noninvasively and continuously administer oral levodopa and carbidopa (LD/CD) into the mouths of patients with Parkinson's Disease (PD). The system is intended to reduce fluctuation in plasma levodopa levels and the associated motor symptoms. The objective of this study is to evaluate the laboratory performance of the DopaFuse Delivery System.

Background

DopaFuse is the first of a new class of drug delivery devices that reside in the mouth and that non-invasively and continuously deliver drugs to patients at a constant rate for absorption via the gastro-intestinal and/or buccal routes. The System consists of a reusable custom dental retainer, its case, and a pre-filled, single-use drug container, shown in Figures 1-3.

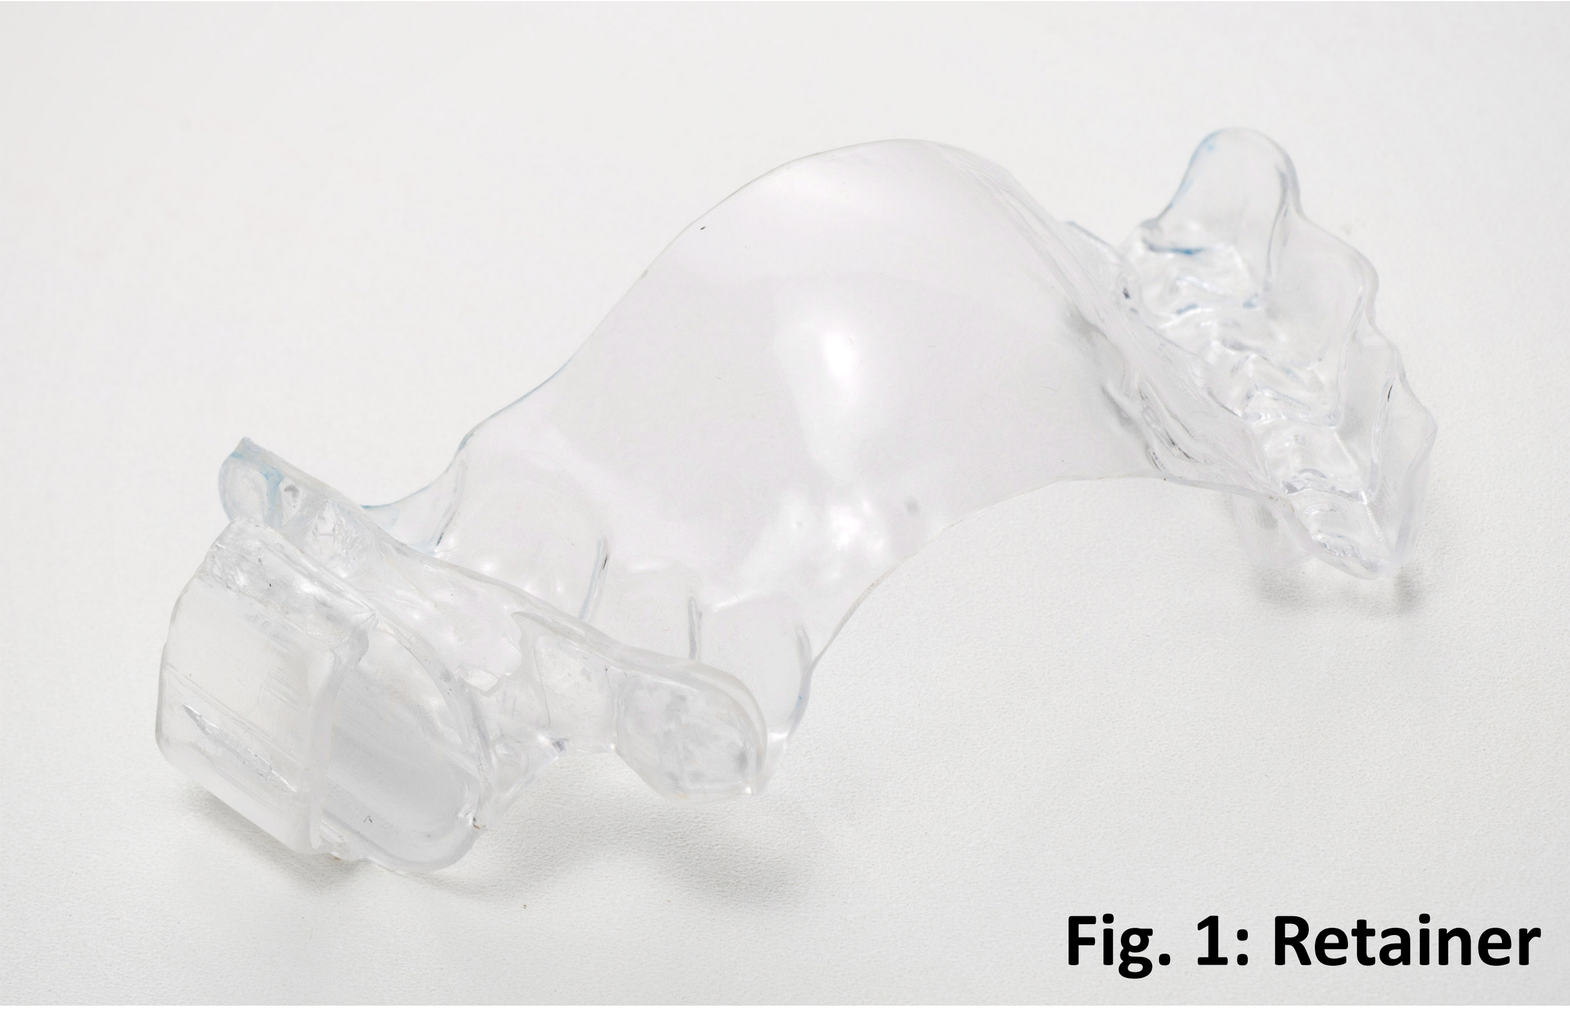

Fig. 1: Retainer

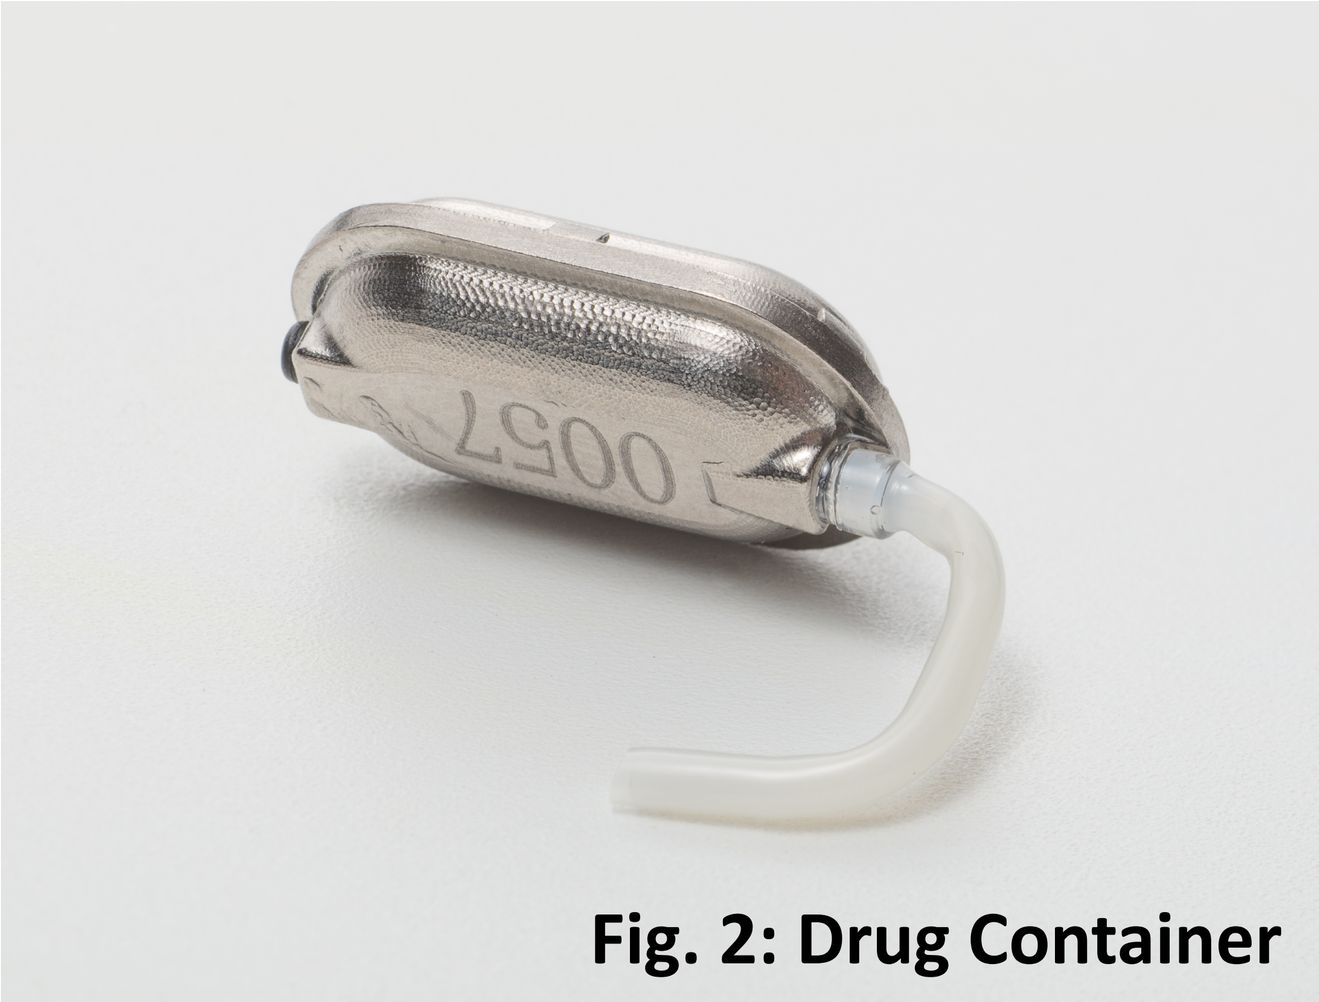
0057

Fig. 2: Drug Container

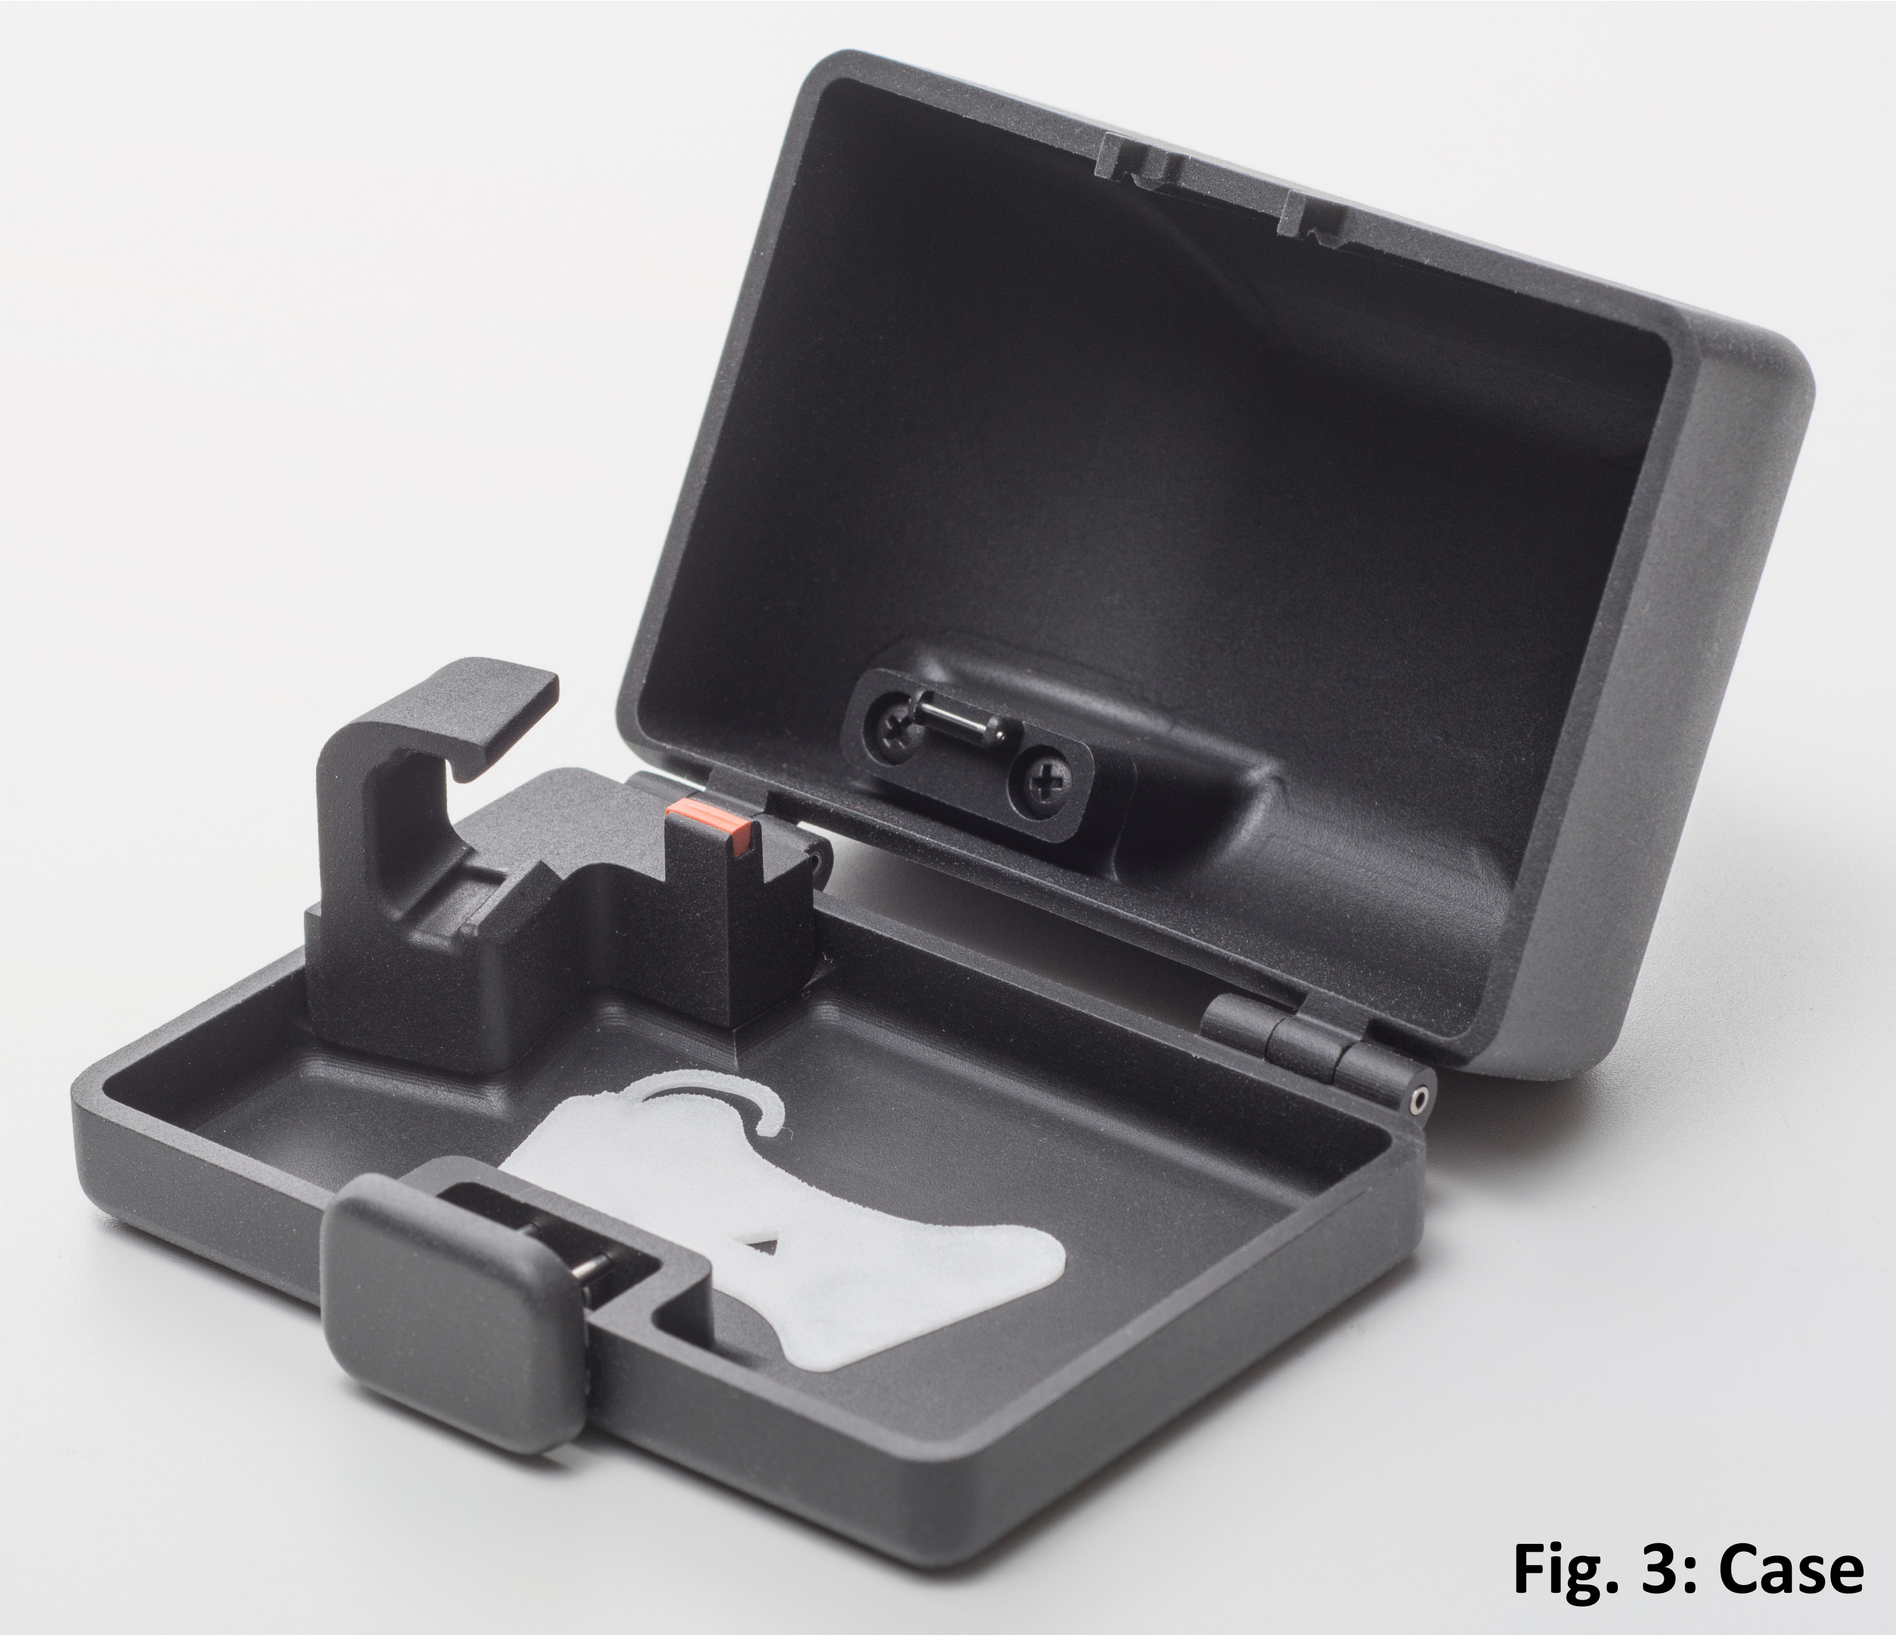

Fig. 3: Case

Multiple clinical studies have demonstrated that continuous administration of a short-acting dopaminergic agents such as levodopa is associated with reduced motor complications in comparison to intermittent dosing with the same agent[1-11]. Continuous intra-intestinal infusion of levodopa has been shown to reduce motor "off" time and increase "on" time with no increase in dyskinesia in comparison to optimized treatment with standard doses of oral levodopa therapy[10, 12,13]. However, intestinal infusion is cumbersome and is associated with frequent and potentially serious adverse events related to the surgical procedure, the tubing and the pump.

SynAgile conducted a single proof-of-concept study in 2015 to test the hypothesis that continuous oral delivery of LD/CD results in improved LD pharmacokinetics and reduced Off time. The study found significantly less variability in plasma LD concentration and reduced Off with continuous versus intermittent oral levodopa delivery.[14] Based on the positive results of the study, the DopaFuse Delivery System is being developed to provide the benefits of continuous LD/CD delivery with fewer complications than existing treatments for advanced PD.

How it Works

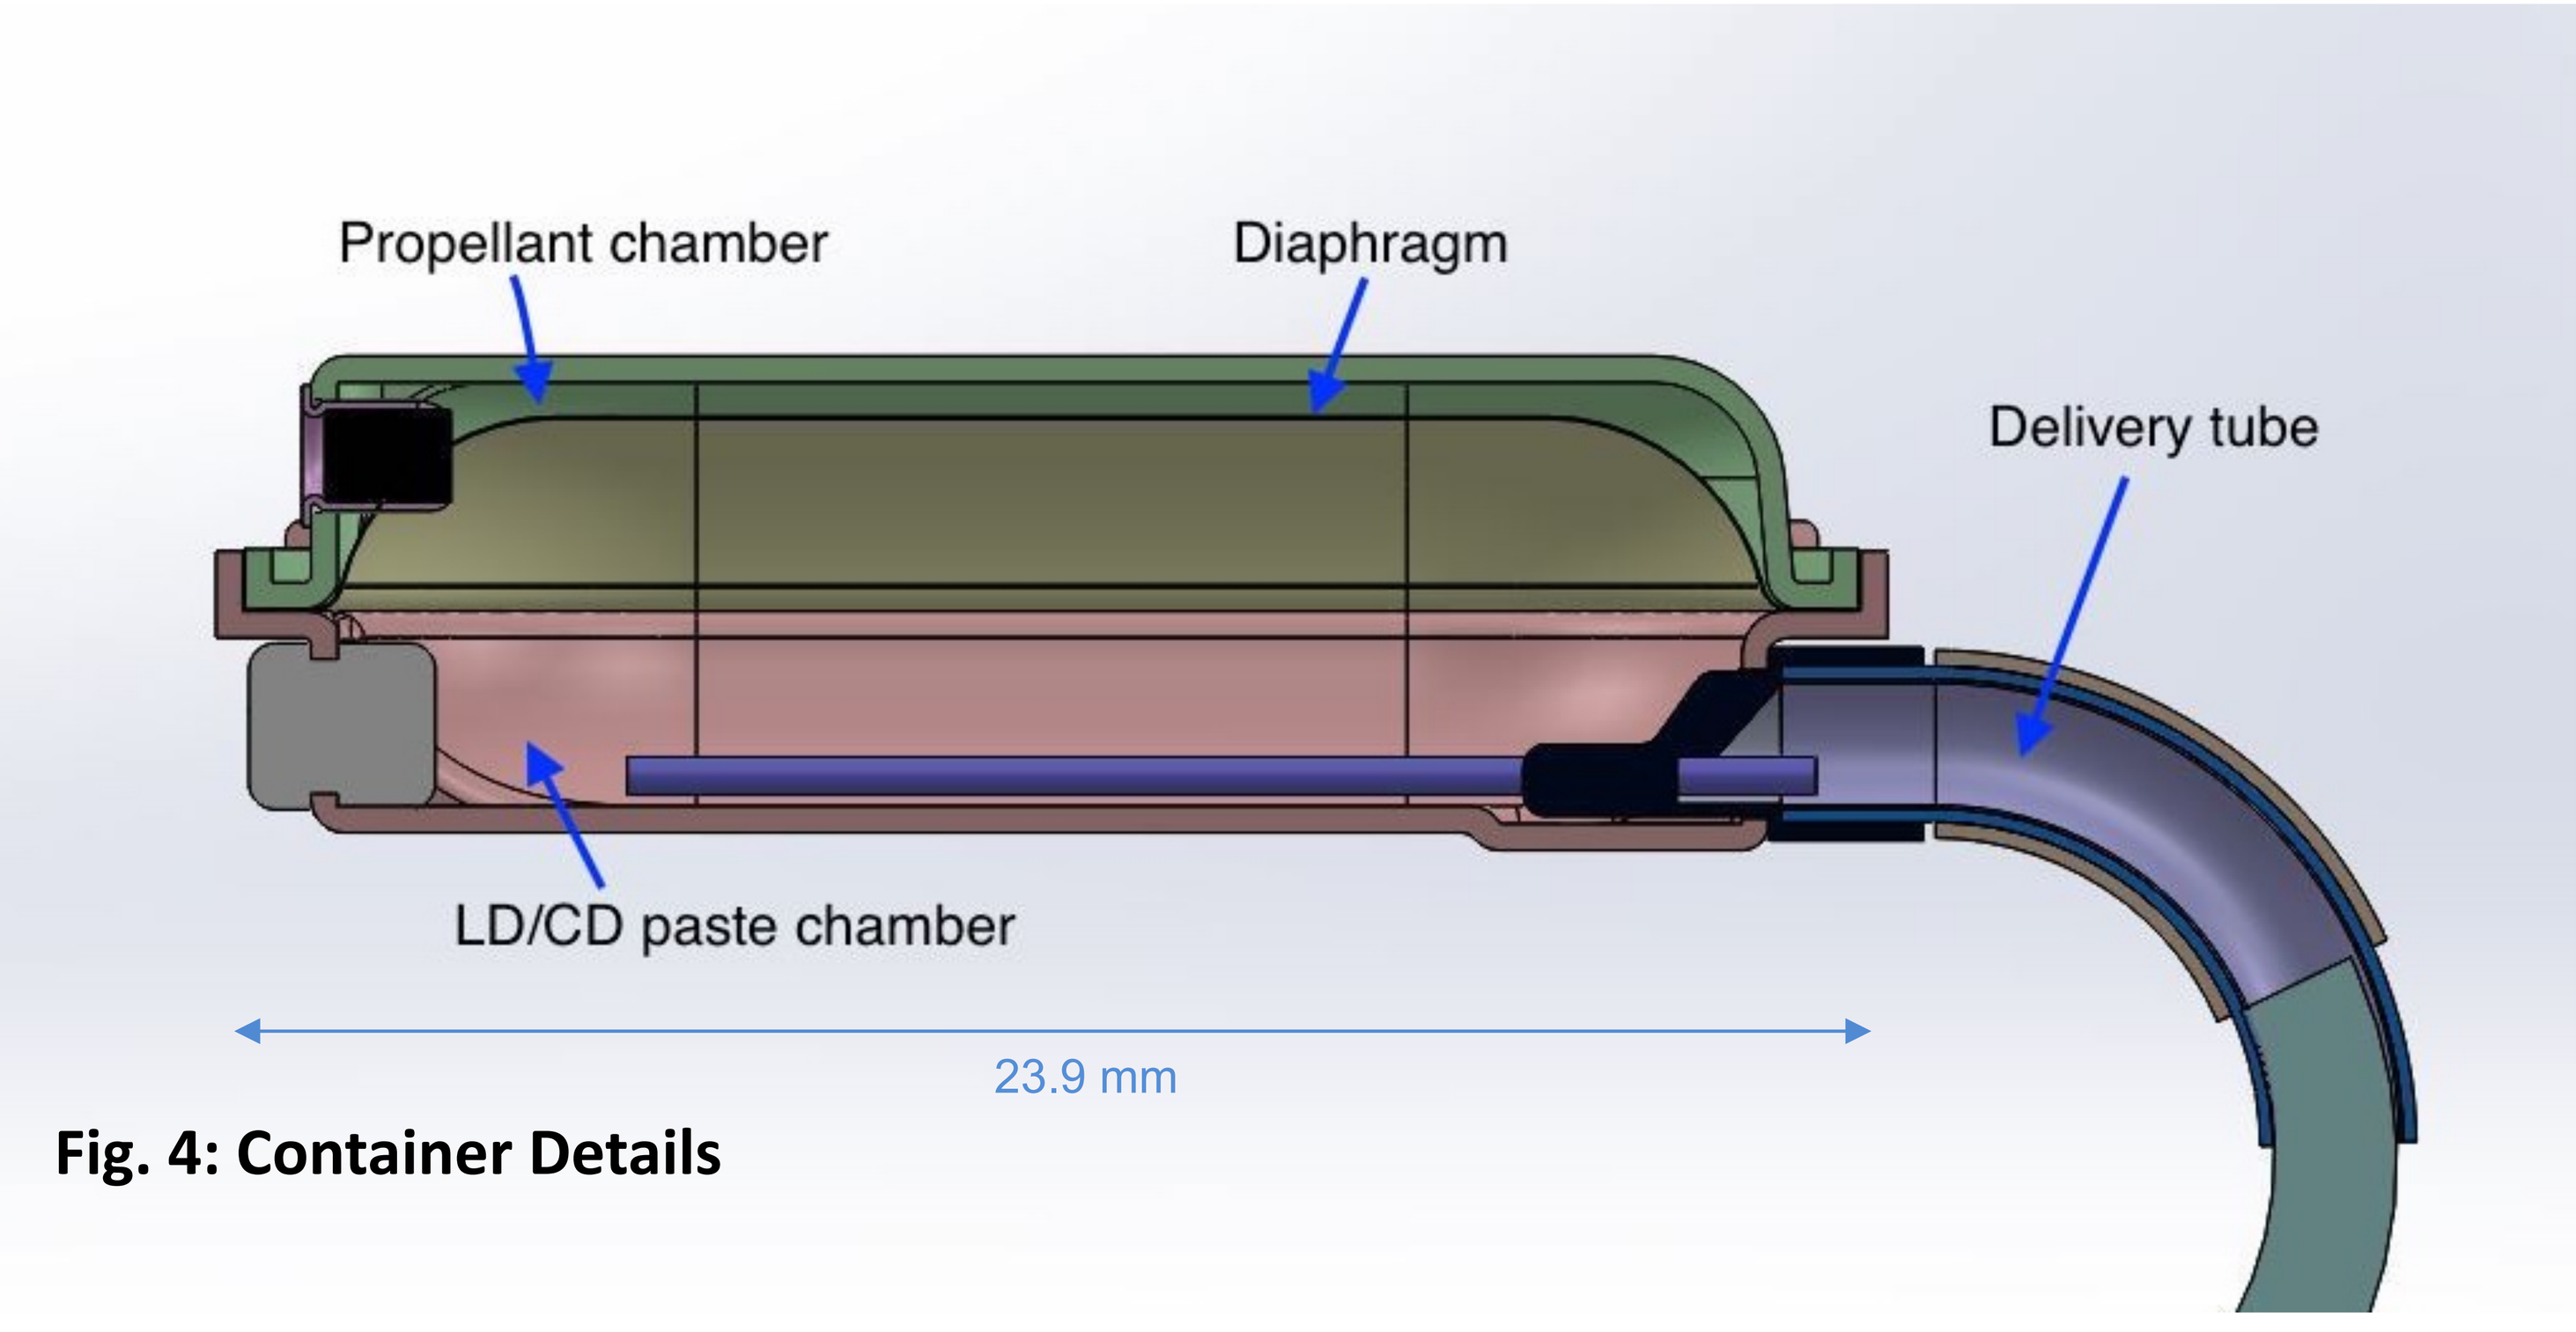
Propellant chamber
Diaphragm
Delivery tube
LD/CD paste chamber
23.9 mm

Fig. 4: Container Details

The drug is a reformulation of LD/CD into a highly concentrated, stable, oral paste. The container has no moving parts, batteries, or electronics. The container consists of two chambers separated by a deformable metal diaphragm, as shown in Figure 4. A propellant (the same as is used in metered dose inhalers) exerts a constant pressure on the flexible diaphragm, which in turn exerts a constant pressure on the drug paste. The drug paste is forced out of the drug chamber through two flow-controlling nozzles and into the delivery tube. The delivery tube wraps around the rearmost tooth and delivers the paste near the back of the tongue. The tasteless LD/CD oral paste disperses in saliva and is swallowed, then absorbed in the gastrointestinal tract.

The container is designed to deliver drug in a continuous, linear fashion within ±20% of the nominal delivery rate for 4-6 hours, depending on the delivery rate. The container is held in place by an oral retainer. The retainer is custom-made for each patient using standard, commercially-available, thermoform retainer materials and molding processes used in dental labs.

The DopaFuse product will be available in multiple LD/CD flow rates to meet the needs of patients with different daily LD/CD dosages. This study tested pumps designed to deliver LD/CD at a rate 68/17 mg/hr, which provides a 12-hour dose of 800/200 mg LD/CD.

Methods

A study of 31 containers was conducted to assess flow rate variability. To test the flow, the delivery tube plug is removed and the paste begins to flow. The weight of the delivered paste is measured hourly. It takes a few minutes for the tube to fill with paste before the paste exits the delivery tube.

Results

The results show that after an initial ramp up period, the DopaFuse Paste is delivered at a constant rate over four hours. The first timepoint is slightly lower due to the time it takes to prime the delivery tube. The average flow rate was 64.7 (SD= 6.2, CI=[52.5, 76.8]).

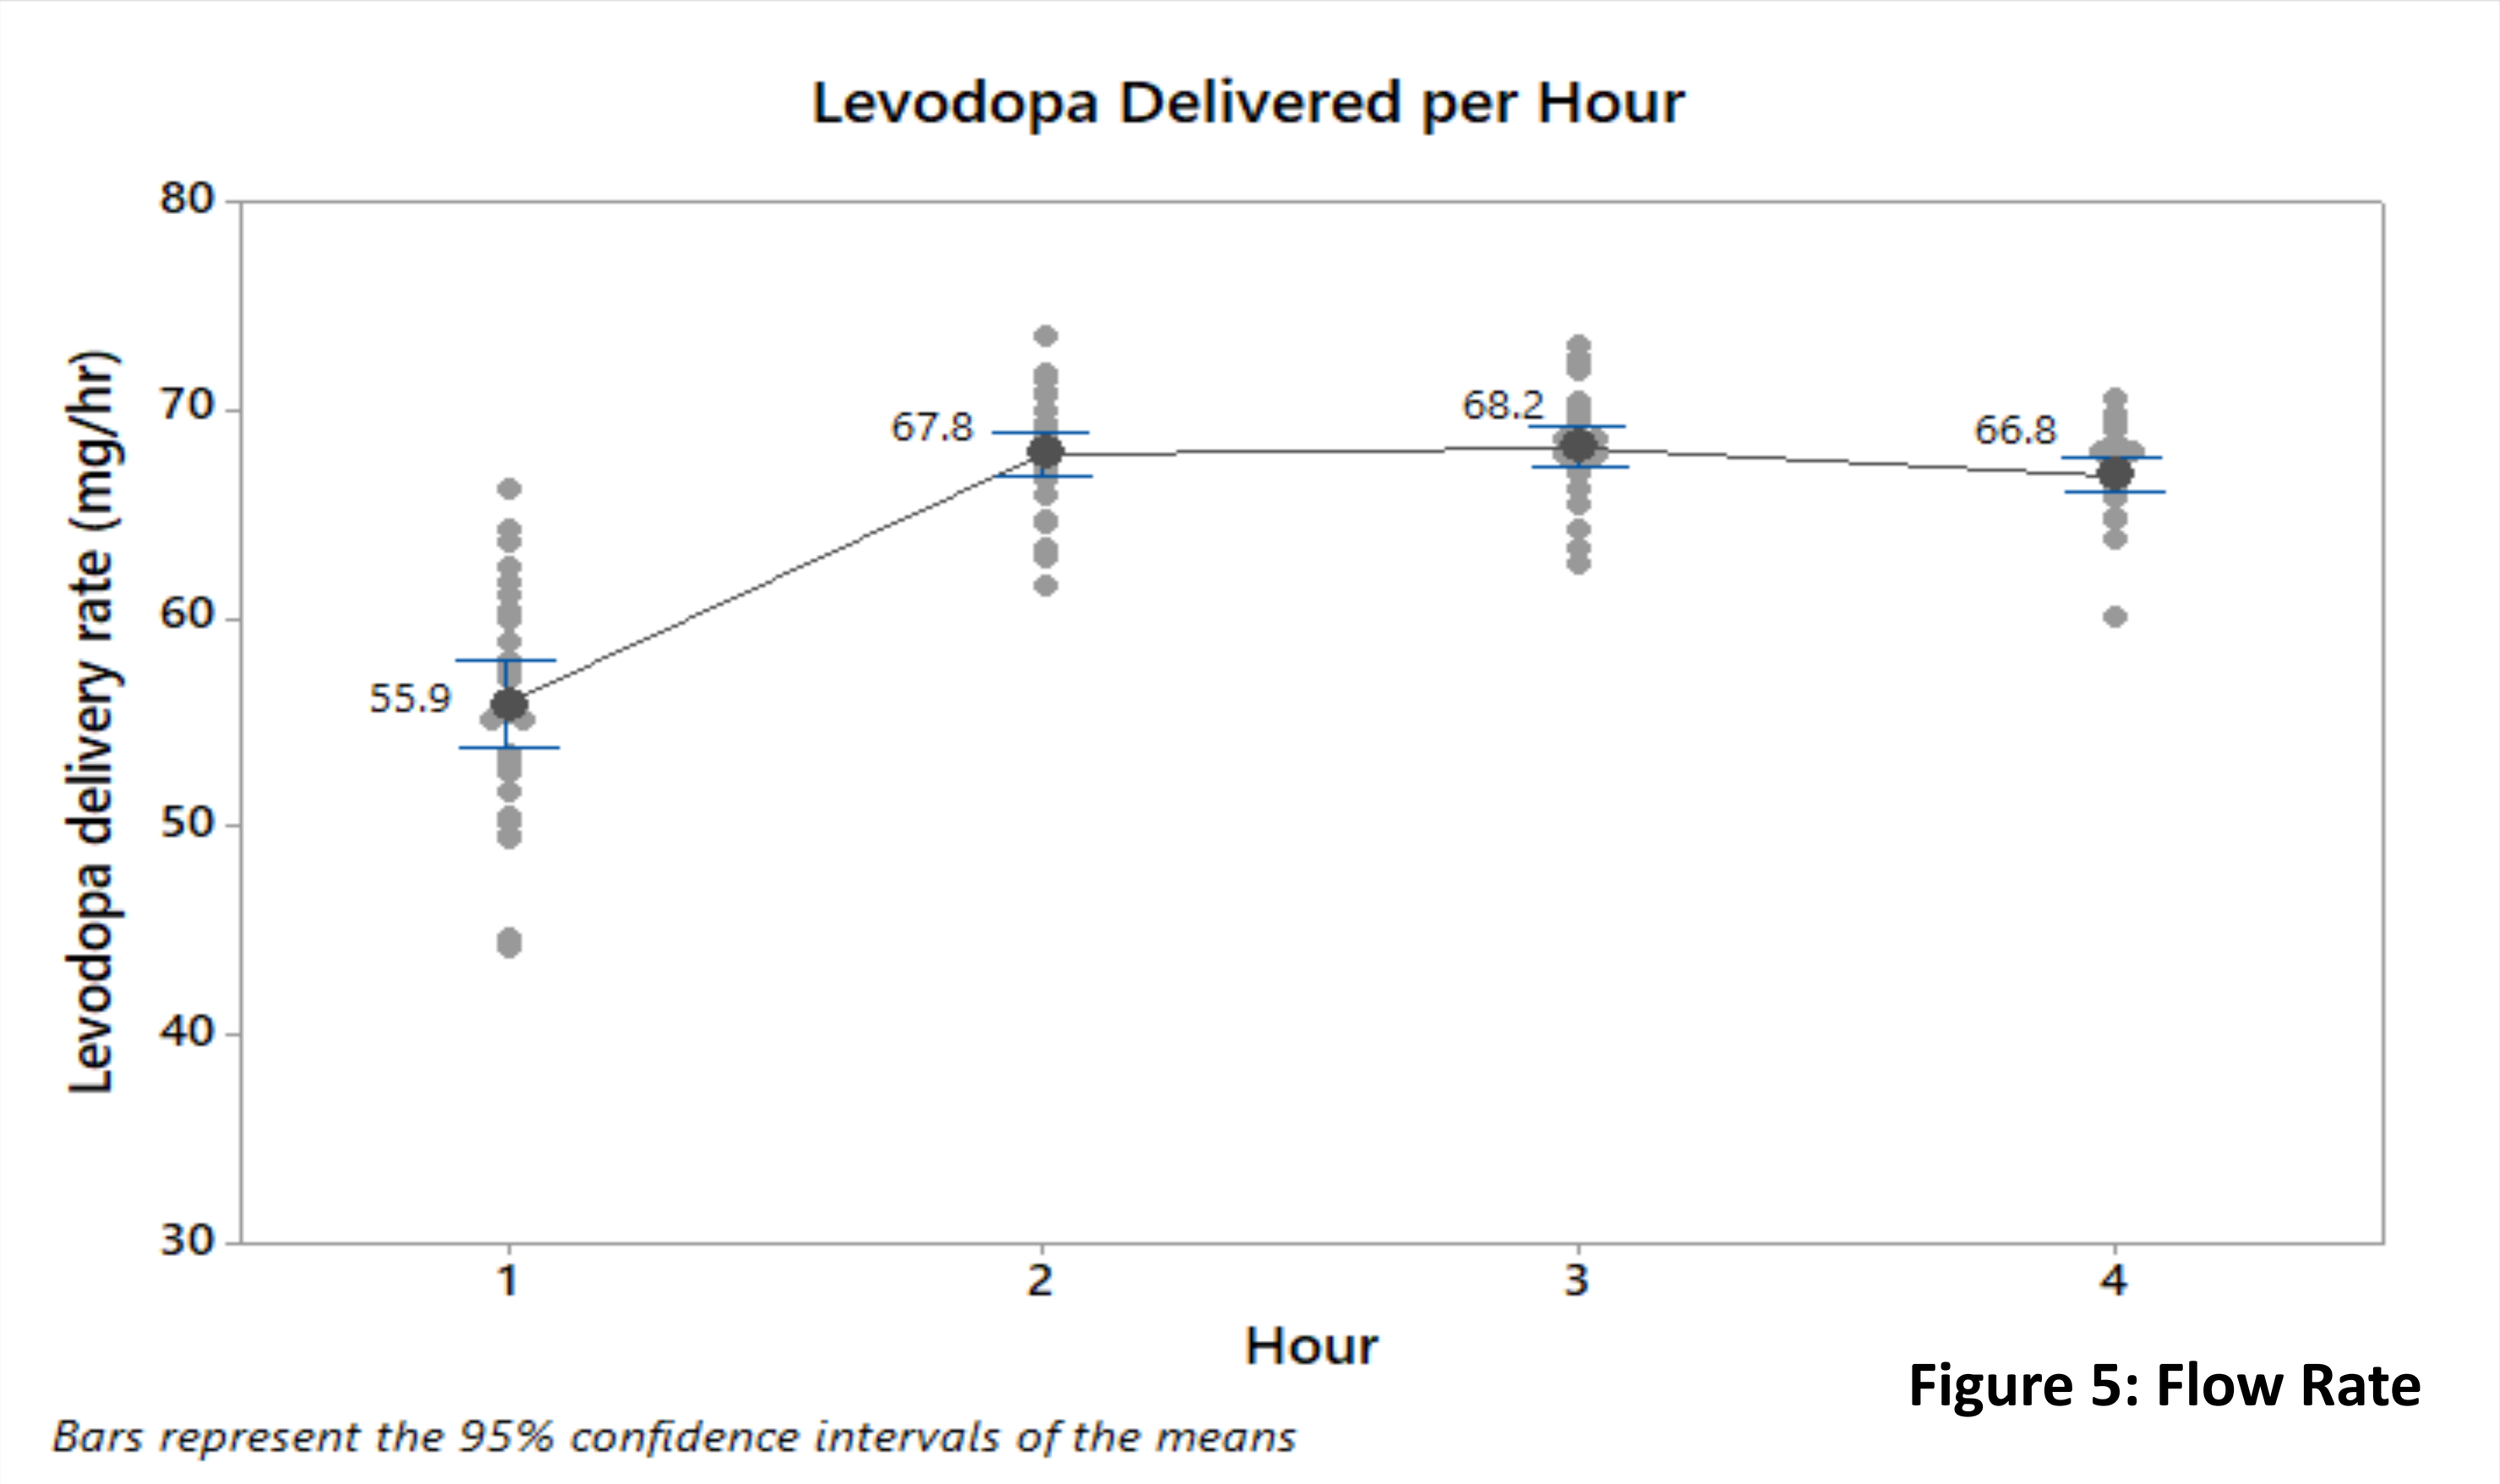
Levodopa Delivered per Hour
Levodopa delivery rate (mg/hr)
80
70
60
50
40
30
55.9
67.8
68.2
66.8
1
2
3
4
Hour
Bars represent the 95% confidence intervals of the means

Figure 5: Flow Rate

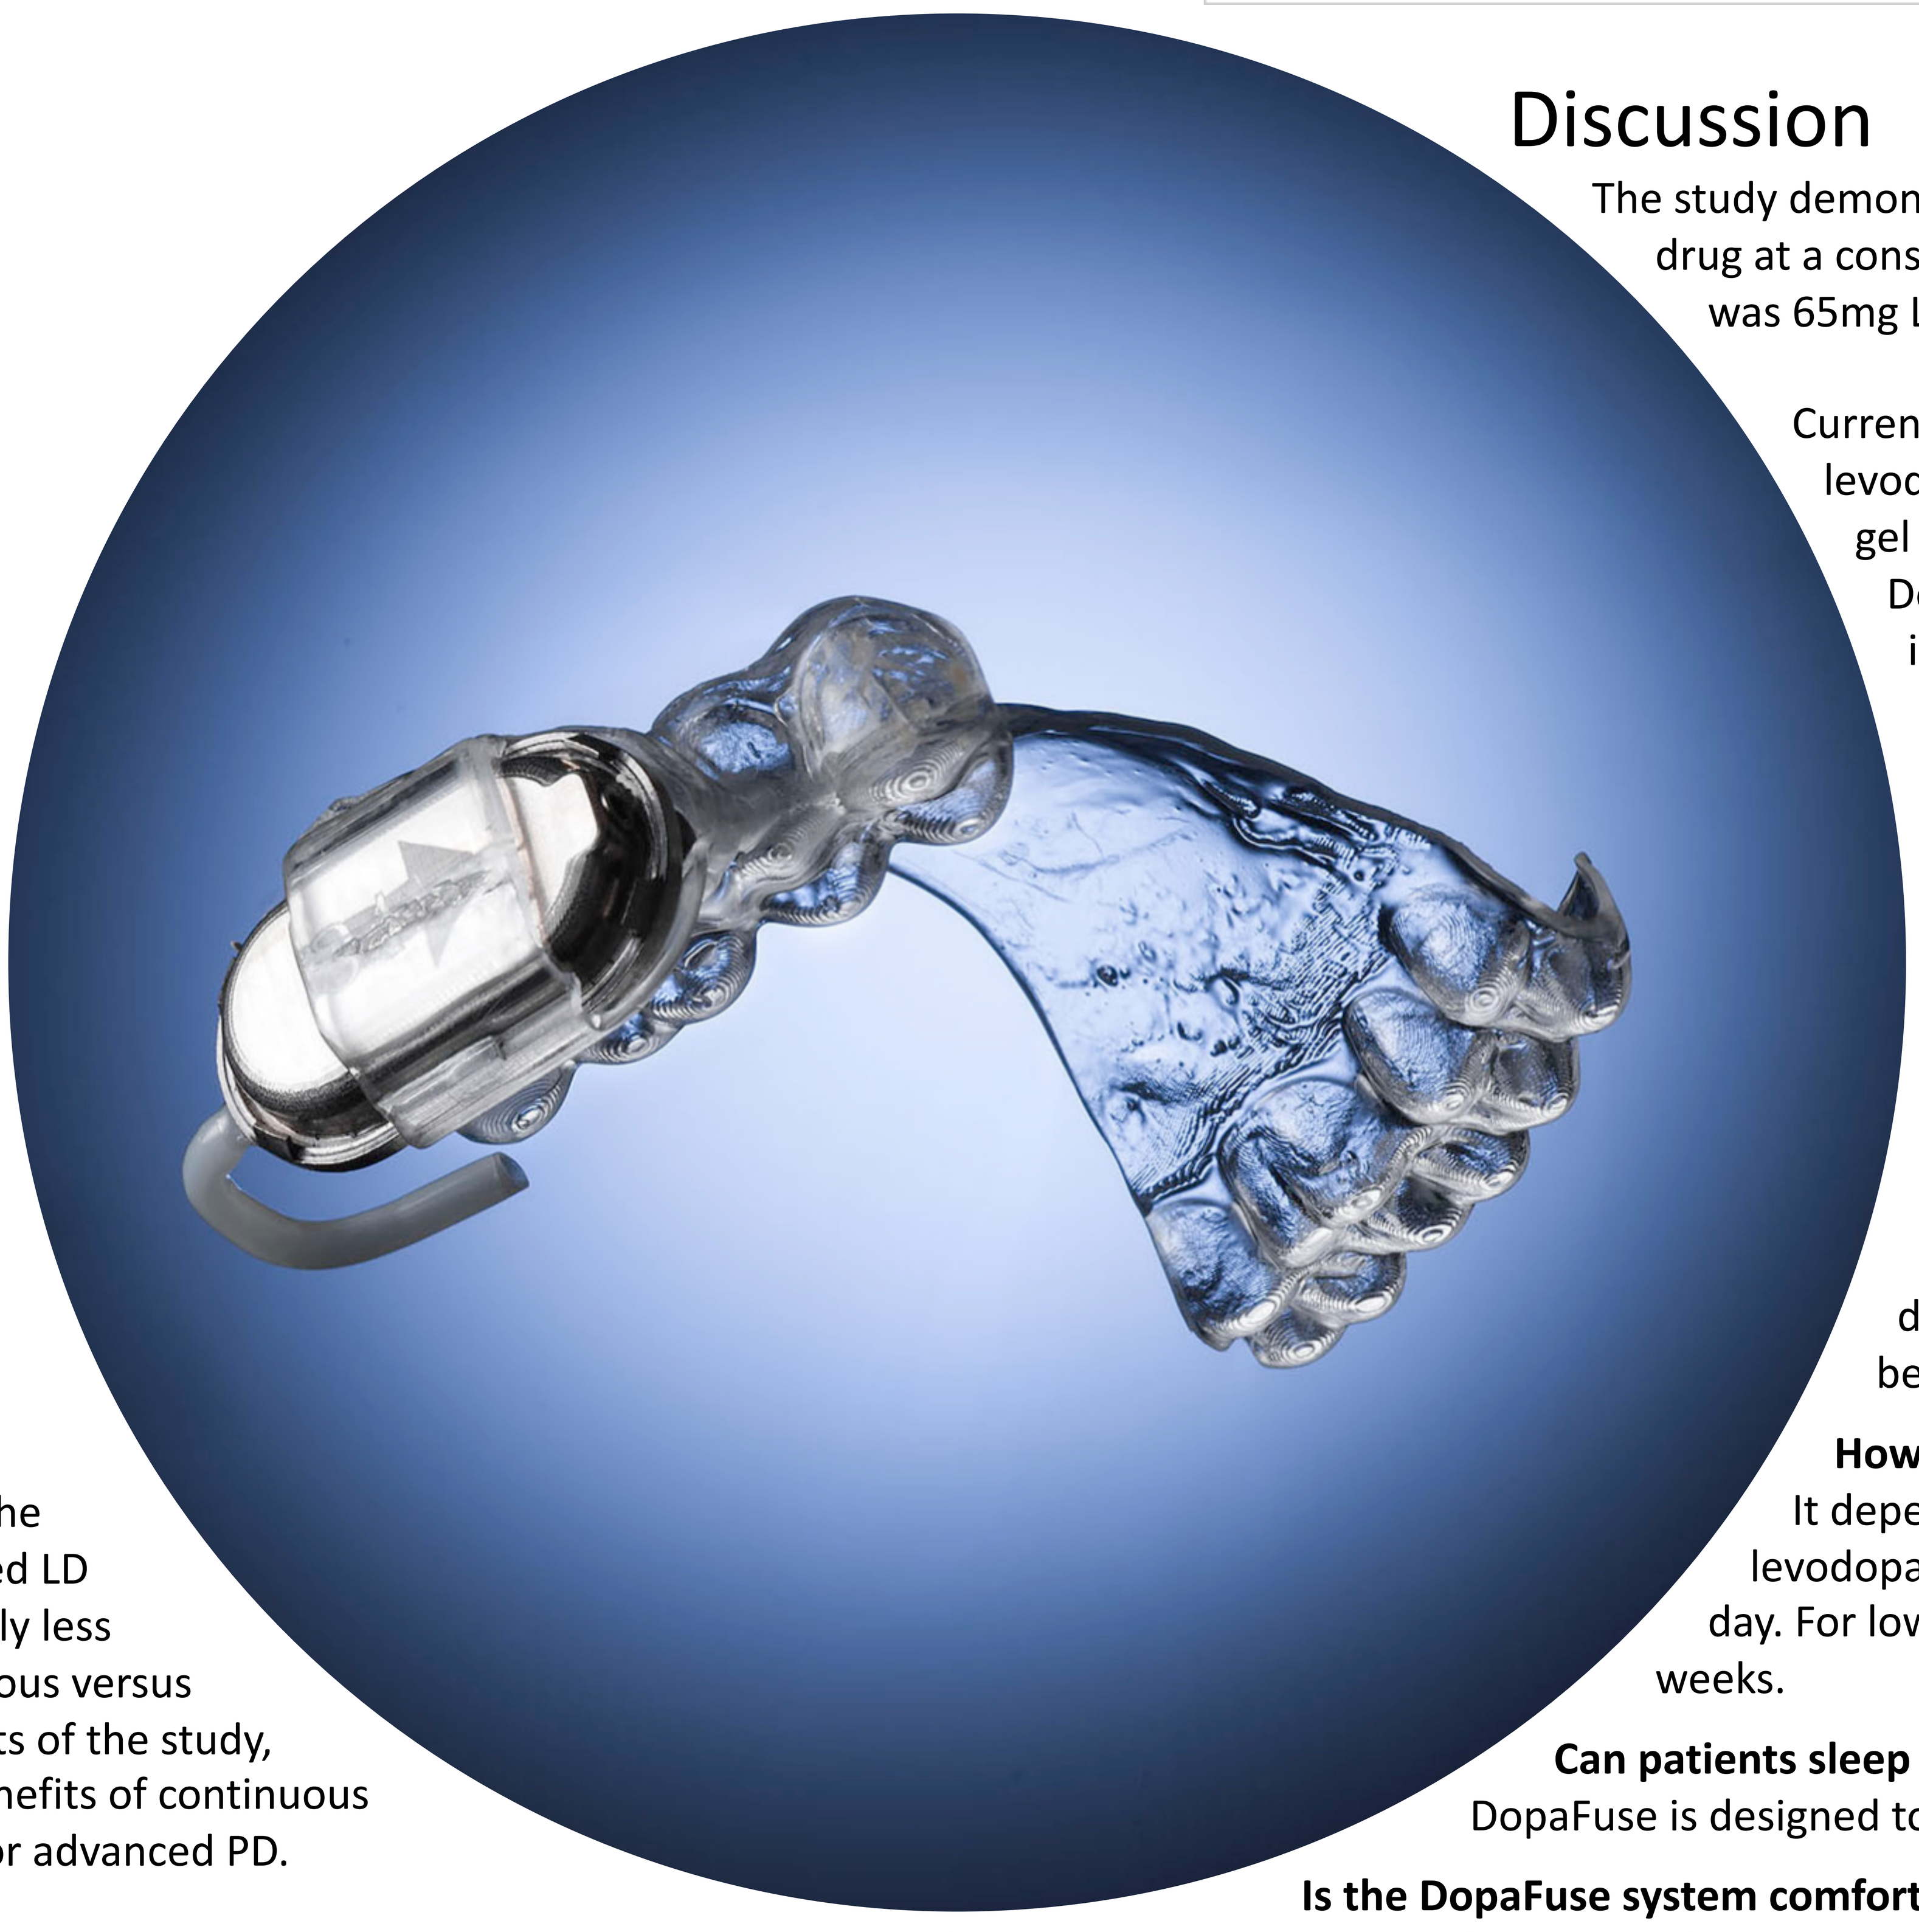

Discussion

The study demonstrates that the DopaFuse container is able to deliver drug at a consistent flow rate for four hours. The average flow rate was 65mg LD/hr for 4 hours.

Currently, the only FDA-approved method for continuous levodopa delivery is a levodopa carbidopa intestinal gel (LCIG) (Duopa®, Duodopa®). This study shows that the DopaFuse Delivery System should be able to provide non-invasive, continuous levodopa without the need for surgical implantation of a pump. DopaFuse products with multiple flow rates are under development to meet the needs of patients who require different dose rates. A Phase 2 pharmacokinetic study of the DopaFuse Delivery System is expected to begin in 2020.

FAQs

Do you taste the drug?
Levodopa and carbidopa have no taste. Most other drugs would be infused so slowly that the user would not be able to perceive any taste.

How long can one drug container last?
It depends on the daily drug dose. For high dose drugs such as levodopa, the drug container should be changed 2-3 times per day. For low dose drugs, one drug container could last up to several weeks.

Can patients sleep with DopaFuse?
DopaFuse is designed to be safe for both daytime and night-time use.

Is the DopaFuse system comfortable?
The DopaFuse system is similar to an ordinary retainer. Retainers are custom made for each patient. Patients will go through a fitting and adjustment with their dentist to ensure comfort.

Can patients eat or drink with DopaFuse?
DopaFuse should be removed for eating. Patients may drink liquids and swallow pills while wearing DopaFuse. Sugary drinks should be avoided while wearing the System.

How do you maintain the System?
The retainer should be cleaned daily with a toothbrush and toothpaste. The case should be cleaned daily with water and a soft cloth.

Is the System visible to others?
The system is designed to be invisible to others.

References

1. Olanow CW, Obeso JA, Stocchi F. Continuous dopamine receptor stimulation in the treatment of Parkinson's Disease: Scientific rationale and clinical implications. Lancet Neurology 2006;5:677-687.
2. Chase TN, Baronti F, Fabbrini G, et al. Rationale for continuous dopaminomimetic therapy of Parkinson's disease. Neurology 1989; 39(Suppl 2):7-10; discussion, 19.
3. Olanow CW, Schapira AH, Rascol O. Continuous dopamine receptor stimulation in early Parkinson's disease. Trends Neurosci 2000;23(10 Suppl):S117-S126.
4. Bibbiani F, Costantini LC, Patel R, et al. Continuous dopaminergic stimulation reduces risk of motor complications in parkinsonian primates. Exp Neurol 2005;192:73-78.
5. Stockwell KA1, Scheller D, Rose S, et al. Continuous administration of rotigotine to MPTP-treated common marmosets enhances anti-parkinsonian activity and reduces dyskinesia induction. Exp Neurol 2009;219:533-542.
6. Stocchi F, Ruggieri S, Vacca L, Olanow CW. Prospective randomized trial of lisuride infusion versus oral levodopa in patients with Parkinson's disease. Brain 2002;125:2058-2066.
7. Kurlan R, Rubin AJ, Miller C, et al.. Duodenal delivery of levodopa for on-off fluctuations in parkinsonism: preliminary observations. Ann Neurol 1986;20: 262-265.
8. Sage JI, Trooskin S, Sonsalla PK, et al. Long-term duodenal infusion of levodopa for motor fluctuations in parkinsonism. Ann Neurol 1988;24:87-89.
9. Bredberg E, Nilsson D, Johansson K, et al. Intraduodenal infusion of a water-based levodopa dispersion for optimisation of the therapeutic effect in severe Parkinson's disease. Eur J Clin Pharmacol 1993;45:117-122.
10. Kurth MC, Tetrud JW, Tanner CM, et al. Double-blind, placebo-controlled, crossover study of duodenal infusion of levodopa/carbidopa in Parkinson's disease patients with 'on-off' fluctuations. Neurology 1993;43:1698-1703.
11. Nyholm D, Askmark H, Gomes-Trolin C, et al. Optimizing levodopa pharmacokinetics: intestinal infusion versus oral sustained-release tablets. Clin Neuropharmacol 2003;26:156-163.
12. Nyholm D, Nilsson Remahl AI, Dizdar N, et al. Duodenal levodopa infusion monotherapy vs oral polypharmacy in advanced Parkinson disease. Neurology 2005;64:216-223.
13. Olanow CW, Kieburtz K, Odin P, et al. Double-Blind, Double-Dummy Study of Continuous Intrajejunal Delivery of Levodopa/Carbidopa Intestinal Gel in Advanced Parkinson's Disease. Lancet Neurol 2014; 13: 141-149.
14. Olanow CW, Torti M, Kieburtz K et al. Continuous oral administration of levodopa in Parkinson's patients with motor fluctuations – a pharmacokinetics, safety, and efficacy study. Mov Disord.2019 Mar;34(3):425-429.

synAgile